Formula X P-01 Manchester

CONTROLLED SUPERSATURATION: ASSESSING THE USE OF EXCIPIENTS IN FORMULATION TO ENHANCE IN VIVO EXPOSURE FOR PRECLINICAL STUDIES

Katerina Vernerova, Richard Taylor
UCB 208 Bath Road – Slough, Berkshire SL1 3WE - United Kingdom
Contact Email: Katerina.Puncochova@ucb.com

Solubility together with permeability are one of the most important parameters to achieve required concentration of the compound in systemic circulation for achieving bioavailability. In combating of poor solubility of drug, the ability to achieve and maintain the supersaturation of drug is substantial. The supersaturated concentration of compound is significantly above its intrinsic solubility. Thus, supersaturation presents the driving force for drug absorption to the blood circulation. The achievement and the maintenance of supersaturation is crucial and offers an opportunity for formulation experts to significantly enhance exposure by maintaining the solubility and prolonging the duration of the supersaturation phase.

Several excipients have been shown to be effective in this context (see Figure 1), however their effect is highly dependent on the compound. This poster describes a new screening approach of determination of the extent and duration of supersaturation in the presence of various excipients using a solvent quench method. This gives the formulation expert precise control over the pH and drug: excipient ratio for each experiment allowing rapid identification of the most effective excipient using a low amount of compound in the formulation development process. Moreover, selected excipients were used in vehicles in preparation of microsuspensions and further tested *in vitro*. Particle size distribution and dissolution rate were determined to support the decision of excipients used *in vivo* studies.

The supersaturation study provides the useful information about the behavior of compound in solution, which replicates fluids in GI track, in a presence of different excipients and can significantly affect the exposure of compound.

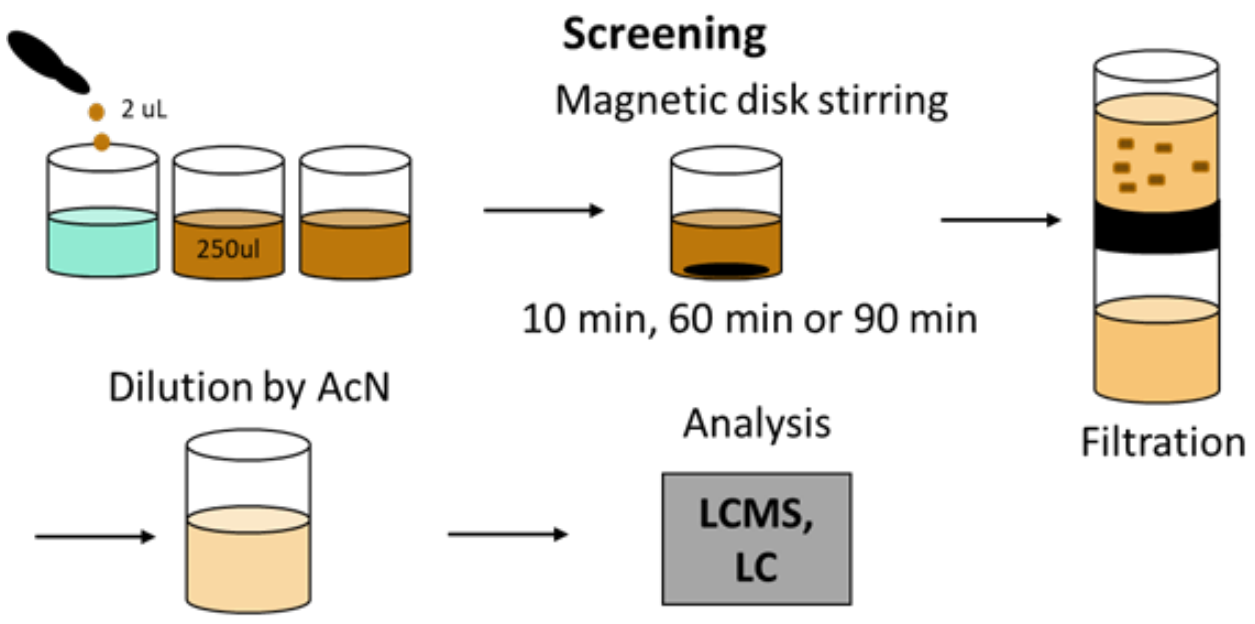
2 uL
250ul
Screening
Magnetic disk stirring
10 min, 60 min or 90 min
Filtration
Dilution by AcN
Analysis
LCMS, LC

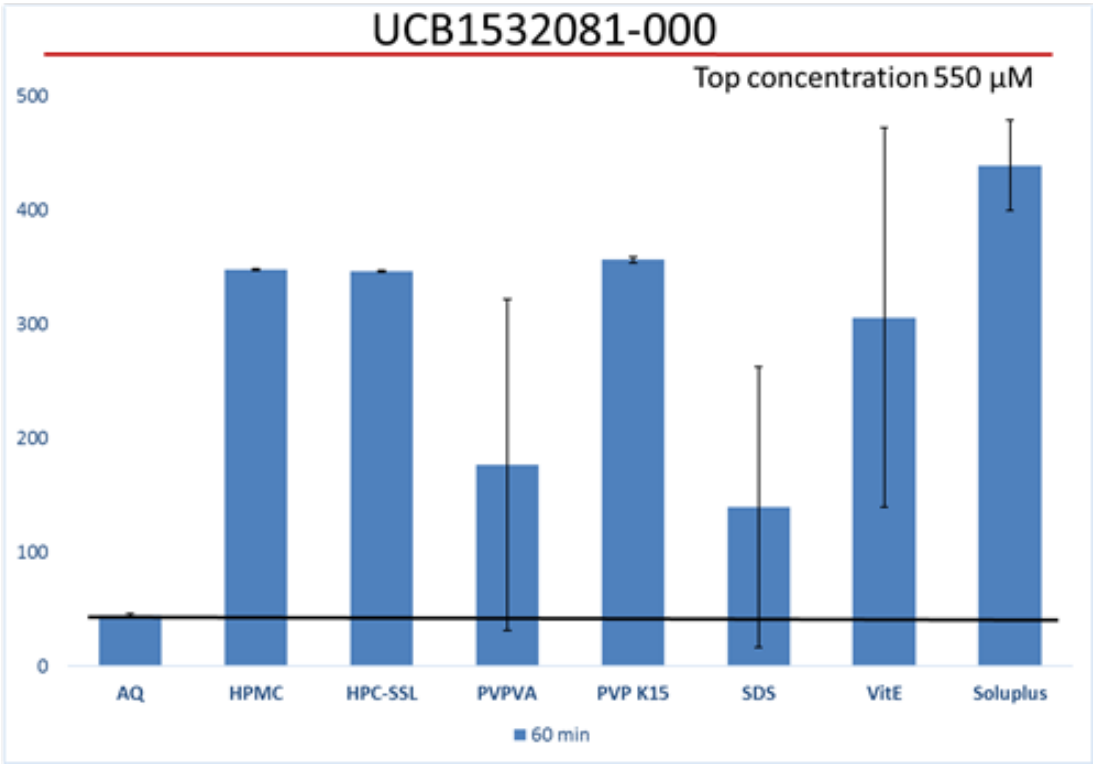
UCB1532081-000
Top concentration 550 µM
Concentration (µM)
0
100
200
300
400
500
AQ
HPMC
HPC-SSL
PVPVA
PVP K15
SDS
VitE
Soluplus
60 min

Figure 1 – Schema of Supersaturation screening and the effect of different excipients on the concentration in the solution for UCB1532081-000.

Back to Programme